PRIMA-102: Predicting biologic and targeted synthetic DMARDs response in rheumatoid arthritis patients using a blood based cell-free DNA_{ac} classifier

Signe Fransen[1], Jason Carlson[1], Bradley Hoolahan[1], Tara Maddala[1], Diana Abdueva[1], Debbie Jakubowski[1], Gordon Lam[2], Alan Kivitz[3], and David Chernoff[1]

[1]Aqtual, Hayward, CA, USA; [2]Arthritis and Osteoporosis Consultants of the Carolinas, Charlotte, NC, USA; [3]Altoona Arthritis & Osteoporosis Center, Duncansville, PA, USA. For questions or trial information, email Signe.Fransen@Aqtual.com.

Background

- Rheumatoid arthritis (RA) presents ongoing challenges in clinical management, particularly for patients with insufficient response or intolerance to targeted therapies.
- Uncontrolled disease results in irreversible joint destruction leading to short and long term disability, diminished quality of life and increased risk of cardiovascular disease, cancer, and death[1].
- The American College of Rheumatology (ACR)[2] and European League Against Rheumatism (EULAR)[3] define the treatment goal as achieving low disease activity using DMARDs*. However, clinicians try several different classes of DMARDS with no guidance on which class the patient is most likely to respond to. This results in chronic cycling of medications (Figure 1).
- This study aims to develop a blood based cell-free DNA classifier to predict therapeutic response to biologic DMARDs (bDMARDs) and targeted synthetic DMARDs (tsDMARDs)(Figure 2).

*conventional synthetic disease modifying anti-rheumatic drugs (cs)DMARD = methotrexate (MTX), leflunomide (LEF), hydroxychloroquine sulfate (HCQ), and sulfasalazine (SSZ); bDMARD = TNFi, anti-IL-6, T cell and B cell; ts DMARD = Janus Kinase Inhibitor (JAKi).

Figure 1. Current Clinical Paradigm

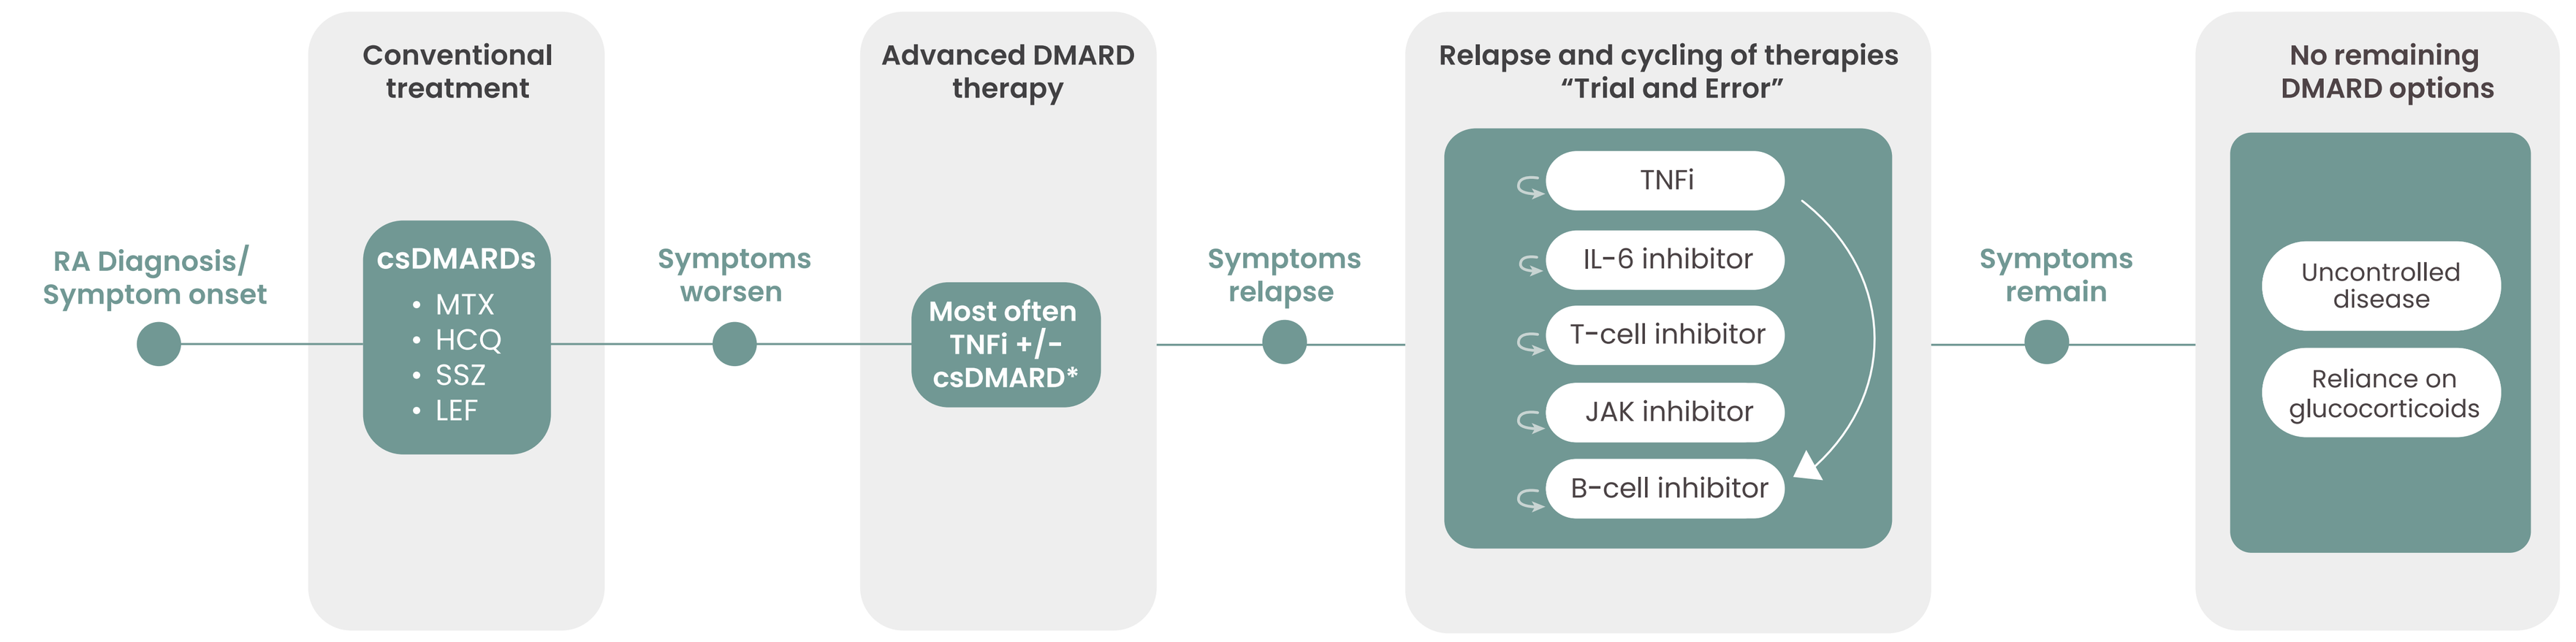
Conventional treatment
Advanced DMARD therapy
Relapse and cycling of therapies "Trial and Error"
No remaining DMARD options
RA Diagnosis/ Symptom onset
csDMARDs
• MTX
• HCQ
• SSZ
• LEF
Symptoms worsen
Most often TNFi +/- csDMARD*
Symptoms relapse
TNFi
IL-6 inhibitor
T-cell inhibitor
JAK inhibitor
B-cell inhibitor
Symptoms remain
Uncontrolled disease
Reliance on glucocorticoids

*ACR recommends combination csDMARD therapy and combination csDMARD + b/tsDMARD therapy

Methods

Figure 2. Technology

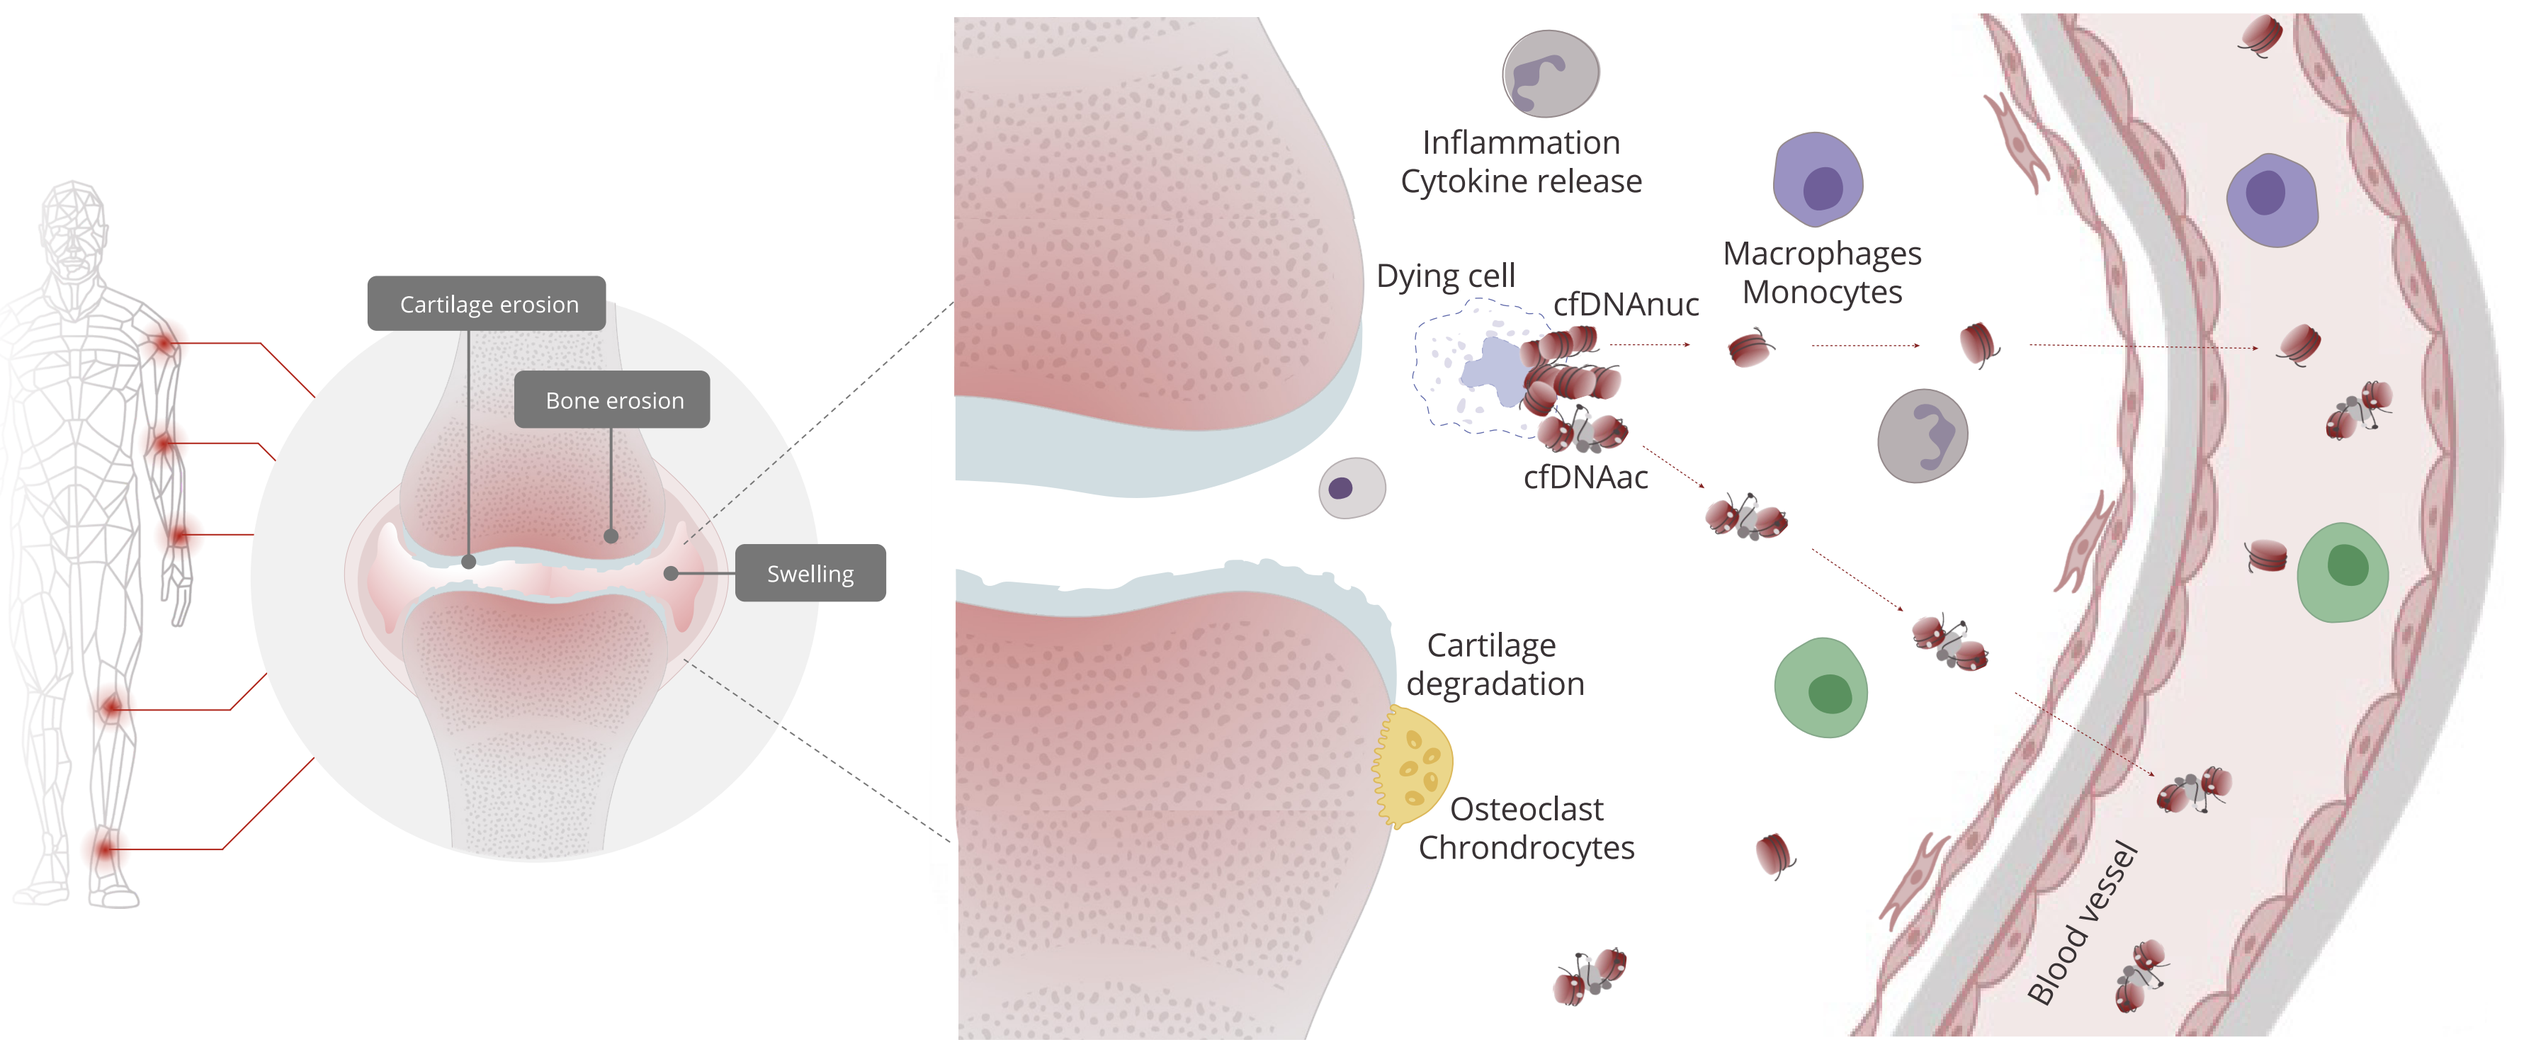
Cartilage erosion
Bone erosion
Swelling
Inflammation
Cytokine release
Dying cell
cfDNAnuc
cfDNAac
Macrophages
Monocytes
Cartilage degradation
Osteoclast
Chrondrocytes
Blood vessel

Background on technology

- Unlike conventional tissue biopsies, cfDNA released into the blood- stream upon cell death provides a non- invasive and dynamic source of molecular information.
- The process of cell death generates two types of cfDNA fragments: 1) conventional nucleosomal fragments ($cfDNA_{nuc}$), and 2) fragments derived from active chromatin ($cfDNA_{ac}$).
- Gene regulation (epigenome) and expression (transcriptome) — in the absence of mRNA — can be assessed by measuring $cfDNA_{ac}$ fragments released into the blood.

Overview of platform

- Aqtual's novel platform provides a non-invasive, real-time assessment of organ- and disease-specific molecular signals.
- cfDNA is extracted from 1 mL of plasma using a proprietary chromatin capture workflow, followed by library preparation, sequencing and machine learning based prediction of therapy response.

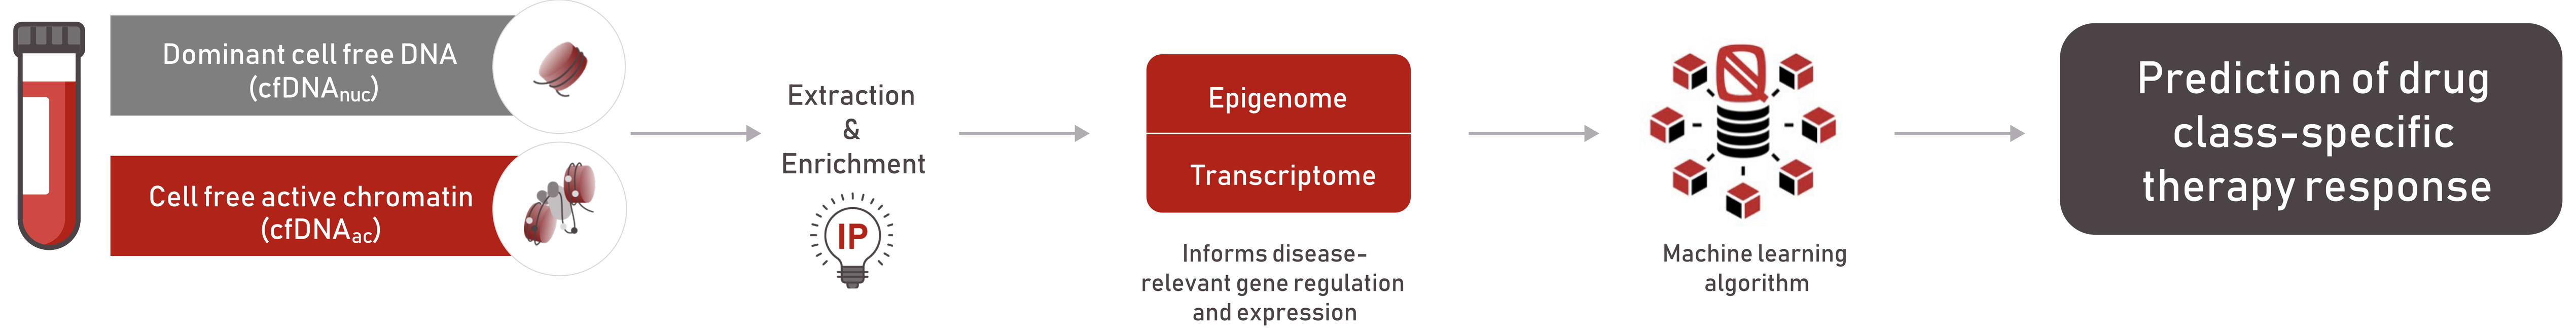
Dominant cell free DNA ($cfDNA_{nuc}$)
Cell free active chromatin ($cfDNA_{ac}$)
Extraction & Enrichment
IP
Epigenome
Transcriptome
Informs disease-relevant gene regulation and expression
Machine learning algorithm
Prediction of drug class-specific therapy response

Study Design

PRIMA 102 is a prospective, multi-center, observational study with blood specimen collection and clinical outcome assessments performed (NCT05936970)

Figure 3. PRIMA 102 Study Schema

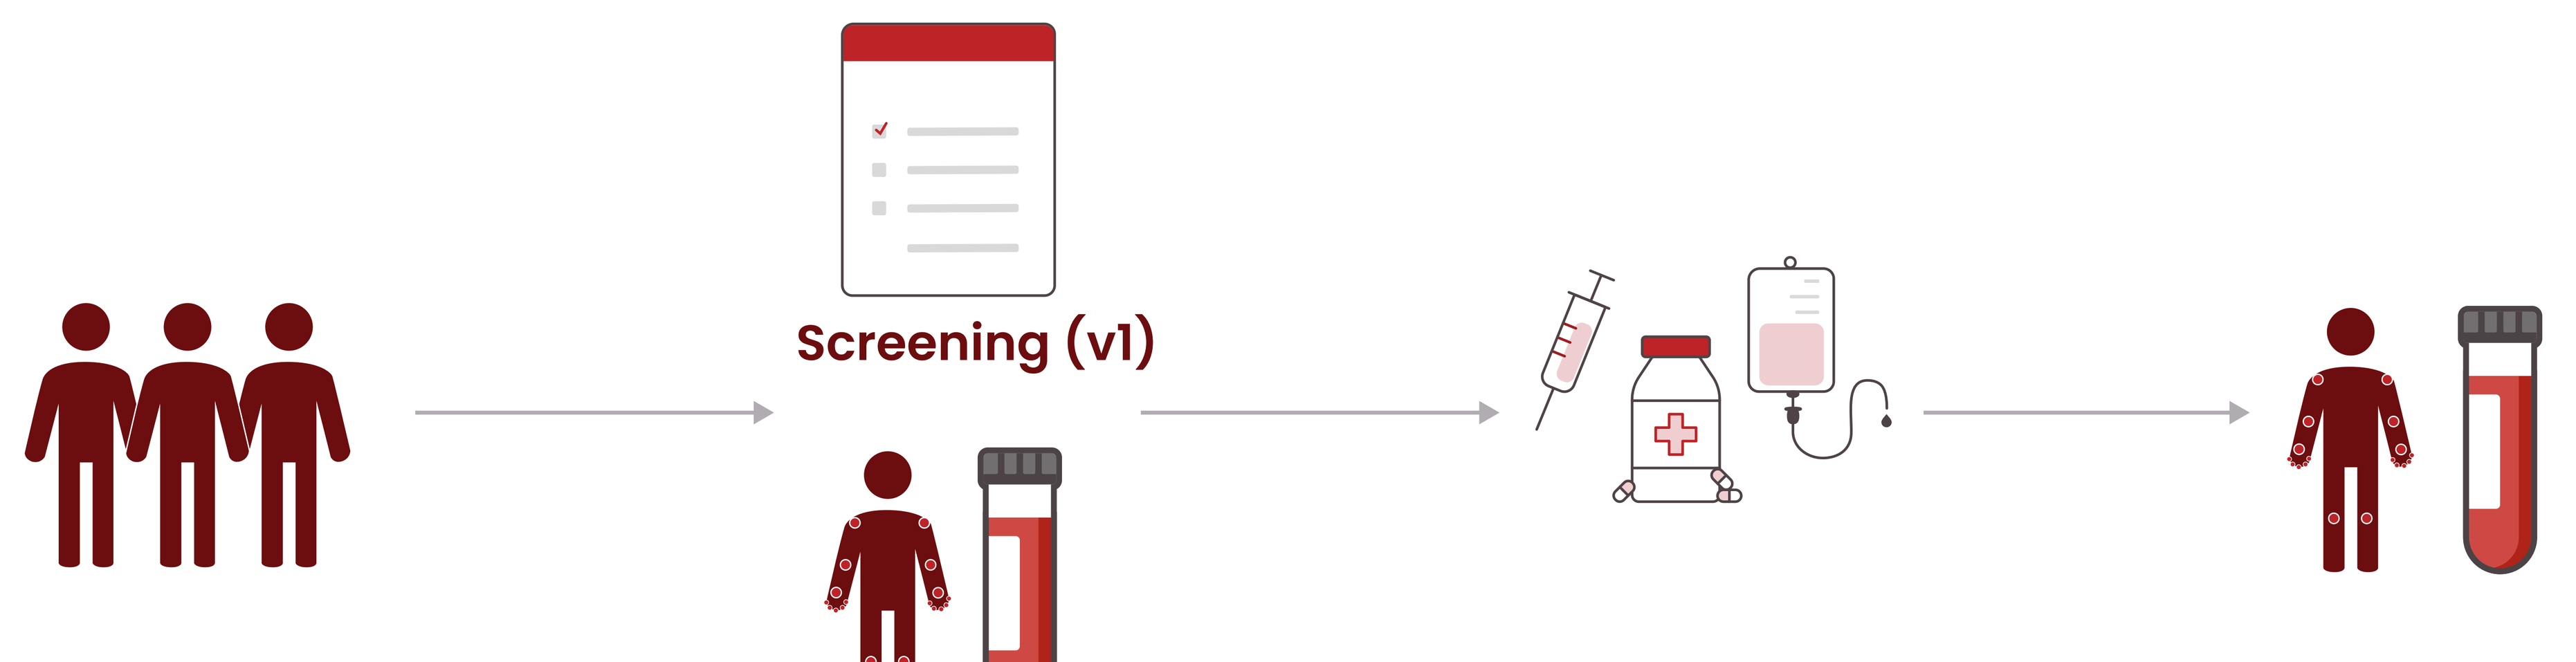
Screening (v1)

Initiation of b- or ts- DMARD planned
- Prior treatment with at least one csDMARD, bDMARD, or tsDMARD

Baseline (v2)[a]
- Informed consent
- Eligibility confirmation
- Blood draw[b]
- Disease Activity Assessments[c]

Initiate new b – or ts-DMARD
- Initiation within 30 days of screening and baseline visits

12 week follow-up (v3)
- Blood draw[b]
- Disease Activity Assessments[c]

V = visit; b/tsDMARD = biologic or targeted synthetic Disease Modifying Anti-Rheumatic Drug; SOC = standard of care
[a] screening and baseline visits should be same day; [b] Includes Aqtual blood test, Rheumatoid Factor (RF), Cyclic Citrullinated Peptide (Anti-CCP), and C-Reactive Protein (CRP); [c] Includes CDAI, DAS-28 CRP, SDAI, EULAR Response, and VAS = visual analog scale disease activity assessment.

Study Objective

The primary objective of this study is to discover features and train classifiers that are predictive of clinical response to bDMARD or tsDMARD treatment.

Study Endpoints

Primary Endpoint
- Change from Baseline in CDAI Score at 12 weeks.

Secondary Endpoints
- Change from Baseline in Disease Activity Score – 28 with C-Reactive Protein (DAS-28 CRP) Score at 12 week.
- Change from Baseline in Simple Disease Activity Index (SDAI) Score at 12 weeks.
- European League Against Rheumatism (EULAR) Response at 12 weeks.

Study Population

Inclusion Criteria
- Ability to understand and provide written informed consent.
- Male or female ≥18 years old.
- Diagnosis of rheumatoid arthritis.
- Prior treatment with at least one csDMARD, bDMARD, or tsDMARD.
- Planning to initiate a new bDMARD (+/- csDMARD) or tsDMARD (+/- csDMARD), within 1 month after enrollment.
- Moderate to high CDAI (defined as >10) with a minimum of 4 tender joints and 4 swollen joints at the time of screening.

Exclusion Criteria
- Unsuitable for participation in the opinion of the principal investigator.

Definition of Response: defined according to minimally clinically important differences established for CDAI[4]

Responders
- If high (CDAI >22) at baseline, then responder defined as having a decrease of ≥12 points , or
- If moderate (CDAI > 10 and ≤22) at baseline, then responder defined as having a decrease of ≥6 points, or
- If the CDAI interpretation at 12 weeks is "low disease activity" or "remission" (total score is ≤10).

Non-responders
- Not meeting the criteria for response by CDAI.

Figure 4. PRIMA 102 study locations

- Currently, there are 16 active sites.
- Site selection and enrollment is ongoing with a target enrollment of ~800 participants.

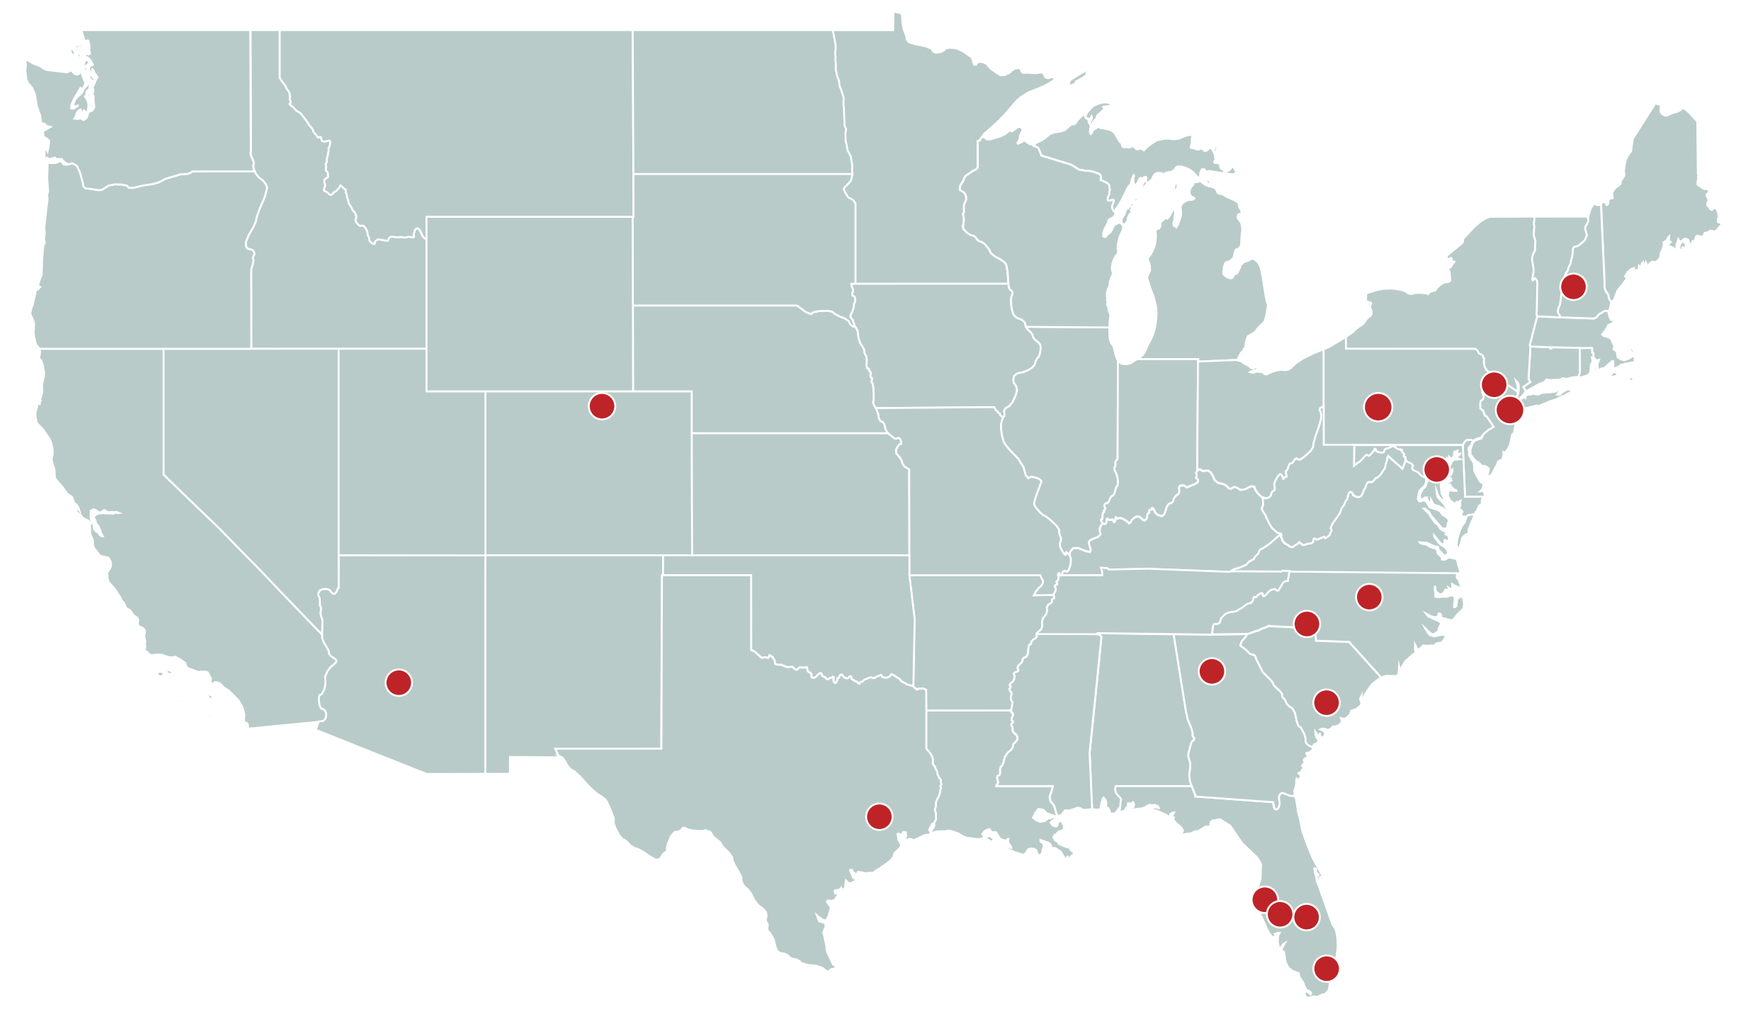

Conclusions

- PRIMA 102 is an ongoing, prospective, multi-center, observational study with blood specimen collection and clinical outcome assessments performed (NCT05936970) with the goal of discovering features and training classifiers that are predictive of clinical response to bDMARD or tsDMARD treatment.
- Cell-free DNA from active chromatin offers a novel approach to capturing molecular features that can predict drug responses in RA patients.
- The development and validation of a blood-based $cfDNA_{ac}$ classifier that delivers a positive prediction of drug-class specific therapy response could guide personalized treatment decisions in RA, improving outcomes for patients who have inadequate response or intolerance to current therapies (Figure 5).

Clinical Implications

Figure 5 . Future clinical paradigm

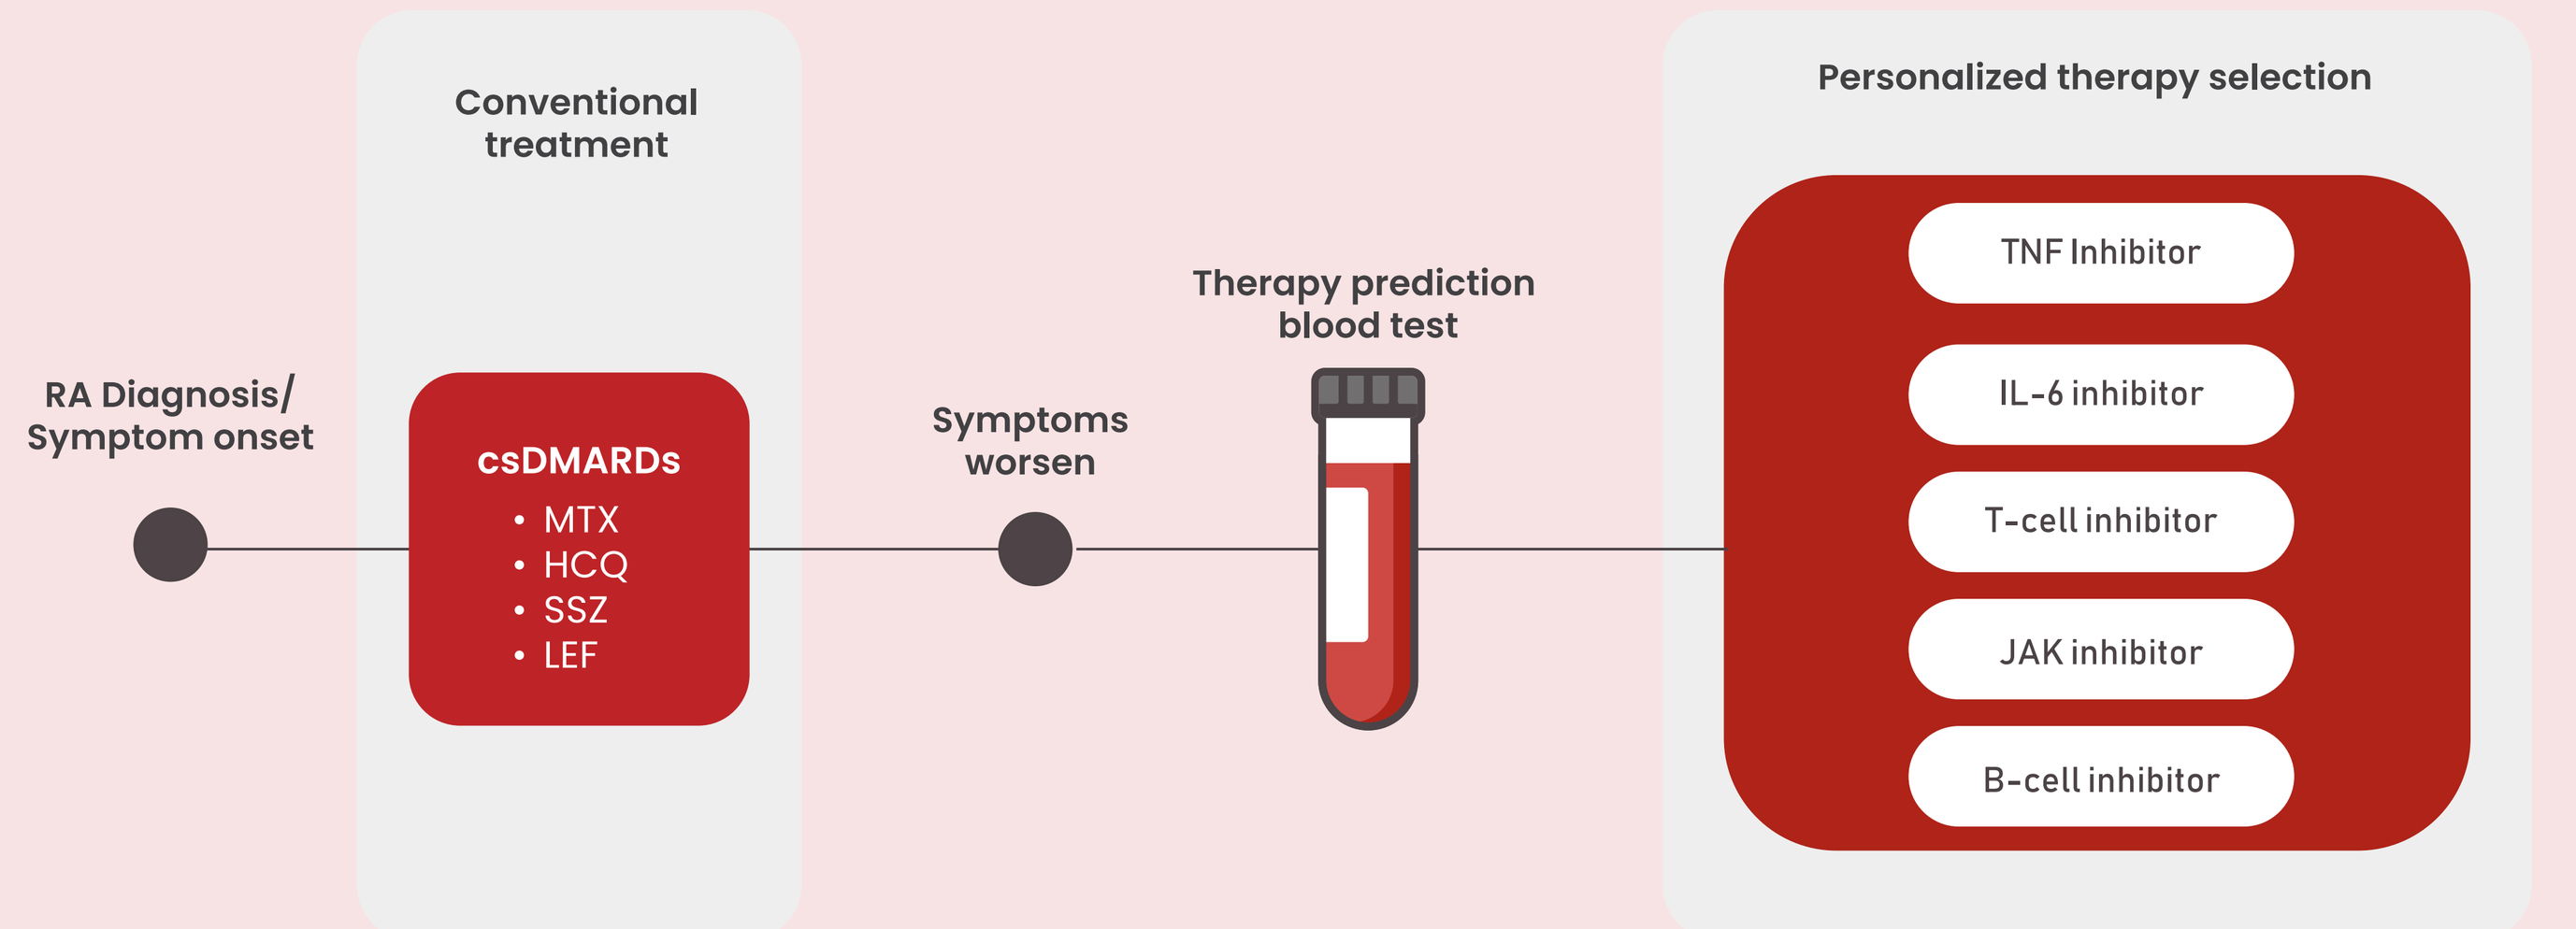
Conventional treatment
Personalized therapy selection
RA Diagnosis/ Symptom onset
csDMARDs
• MTX
• HCQ
• SSZ
• LEF
Symptoms worsen
Therapy prediction blood test
TNF Inhibitor
IL-6 inhibitor
T-cell inhibitor
JAK inhibitor
B-cell inhibitor

The goal of this study is to collect blood and clinical data from RA patients undergoing a switch in therapy. This data will inform the development of a diagnostic test that could enable personalized treatment strategies and improve patient outcomes in RA management.

References

1. www.cdc.gov/arthritis/basics/rheumatoid-arthritis.html
2. Fraenkel, et al., *Arthritis Care Res*. 2021
3. Smolen, et al., *Ann Rheum Dis*. 2020
4. Curtis et al., *Arthritis Care Res*. 2015

Acknowledgements

The authors gratefully acknowledge the Aqtual clinical operations team and all PRIMA 102 participants and principal investigators.

Disclosure Information: SF is the presenter and an employee of Aqtual. DC is the lead investigator and a scientific advisory board member for Aqtual.